POPULATION PHARMACOKINETICS OF DABRAFENIB (GSK2118436), A BRAF INHIBITOR IN DEVELOPMENT FOR THE TREATMENT OF BRAF V600 MUTATION POSITIVE MELANOMA

E Gibiansky [1], A O'Hagan [2], P Haney [2], J Switzky [2], V Goodman[2], D Ouellet[2]

[1]QuantPharm LLC, North Potomac, MD; [2]GlaxoSmithKline, Research Triangle Park, NC and Upper Providence, PA

Introduction

Dabrafenib (GSK2118436) is a potent and selective small-molecule inhibitor of BRAF kinase activity that is currently being developed for the treatment of BRAF V600 mutation-positive tumors. Clinical activity was first observed in a Phase I, first-time-in-human (FTIH) study, with a confirmed response rate of 56% in patients with V600E mutation-positive melanoma (Falchook, 2012). Efficacy of oral dabrafenib 150 mg twice daily was confirmed in BREAK-3, a Phase III study in patients with BRAF V600E mutation-positive melanoma (Hauschild, 2012) as well as in BREAK-MB, a Phase II study in patients with melanoma who had brain metastases (Long, 2012).

Dabrafenib capsules are absorbed rapidly after oral dosing with Tmax of 1.5 hr. Increases in exposure are less than dose-proportional after repeat dosing, as exposure decreases over time due to auto-induction. Dabrafenib is a CYP2C8/CYP3A4 substrate. In early studies, dabrafenib was administered as gelatin capsules, while later clinical studies were conducted using HPMC capsules, which provided higher exposure to dabrafenib.

Objectives

- ❑ To characterize the population PK of dabrafenib;
- ❑ To evaluate the effect of various demographic factors/potential covariates on GSK2118436 pharmacokinetics;
- ❑ To provide posthoc estimates of the individual PK parameters for a subsequent PK/PD evaluation.

Methods

- ❑ Data from 4 studies (Table 1) were pooled for this analysis

Table 1

Study No.	Study Description	Dose	PK Assessments
BRF112680 (FTIH) Study 1	Phase I, Open-Label, Multiple-Dose, Dose-Escalation, study in Subjects with Solid Tumors. Part 1: Dose Escalation Part 2: Cohort Expansion in Subjects with V600 Mutation Positive Tumors	Part 1: 12 mg (12 mg QD) to 600 mg (300 mg BID) (n=107) Part 2: 50 mg BID and 150 mg BID (n=74) (gelatin capsules)	Part 1: Full PK on Day 1 and/or Day 8 or 15 Part 2: Sparse sampling
BRF113710 (Phase 2) Study 2	Phase 2, single-arm, open-label study in subjects with BRAF mutant metastatic melanoma.	Dose of 150 mg BID (N=87) (gelatin capsules)	Predose, and 1-3 post-dose at Wk 3 and AM or PM sample at Wks 6, 9, 12, 20, 28
BRF113929 (Phase 2 with brain mets) Study 3	Phase 2 open-label, two-Cohort, study in treatment naïve and previously treated subjects with BRAF mutation-positive metastatic melanoma to the brain	Dose of 150 mg BID (N=148) (HPMC capsules)	Predose, and 1-3 post-dose at Week 4 and AM or PM sample at Wks 8, 16, 24, 32.
BRF113683 (Phase 3) Study 4	Phase 3, randomized open-label study of dabrafenib vs dacarbazine (DTIC) (3:1 ratio) in subjects with BRAF mutant metastatic melanoma.	Dose of 150 mg BID (N=179) (HPMC capsules)	All: AM or PM sample at Wks 3, 6, 9, 12, 18, 24 Subset: Full PK Wk 6

Methods – Cont.

- ❑ Nonlinear mixed effects modeling approach (NONMEM 7.2) with FOCEI method was used.
- ❑ Test covariates using a full model with the following covariates:

CL/F: Effects of body weight, sex, age groups, mild hepatic impairment, mild and moderate renal impairment, concomitant CYP3A4 inhibitors or inducers and capsule shell;
Vc/F: Effects of body weight and sex;
Vp/F, Q/F: Effect of body weight;
F, Ka: Effect of capsule shell.

- ❑ Population PK models were evaluated using diagnostic plots, bootstrap and visual predictive check (VPC)

Results

- ❑ Dabrafenib concentrations (n=1931) in 595 subjects were pooled; PK profiles shown in Figure 1.
- ❑ PK described by a two-compartment model with first order absorption (Ka), absorption delay (Tlag), central and peripheral volume (Vc/F and Vp/F), distributional clearance (Q/F), and with elimination described by non-inducible apparent clearance (CL_0/F), and an inducible apparent clearance that increased almost linearly with dose and increased with time until it reached steady-state (CLind,ss/F), with a half-life of induction of T_{50}.

Figure 1

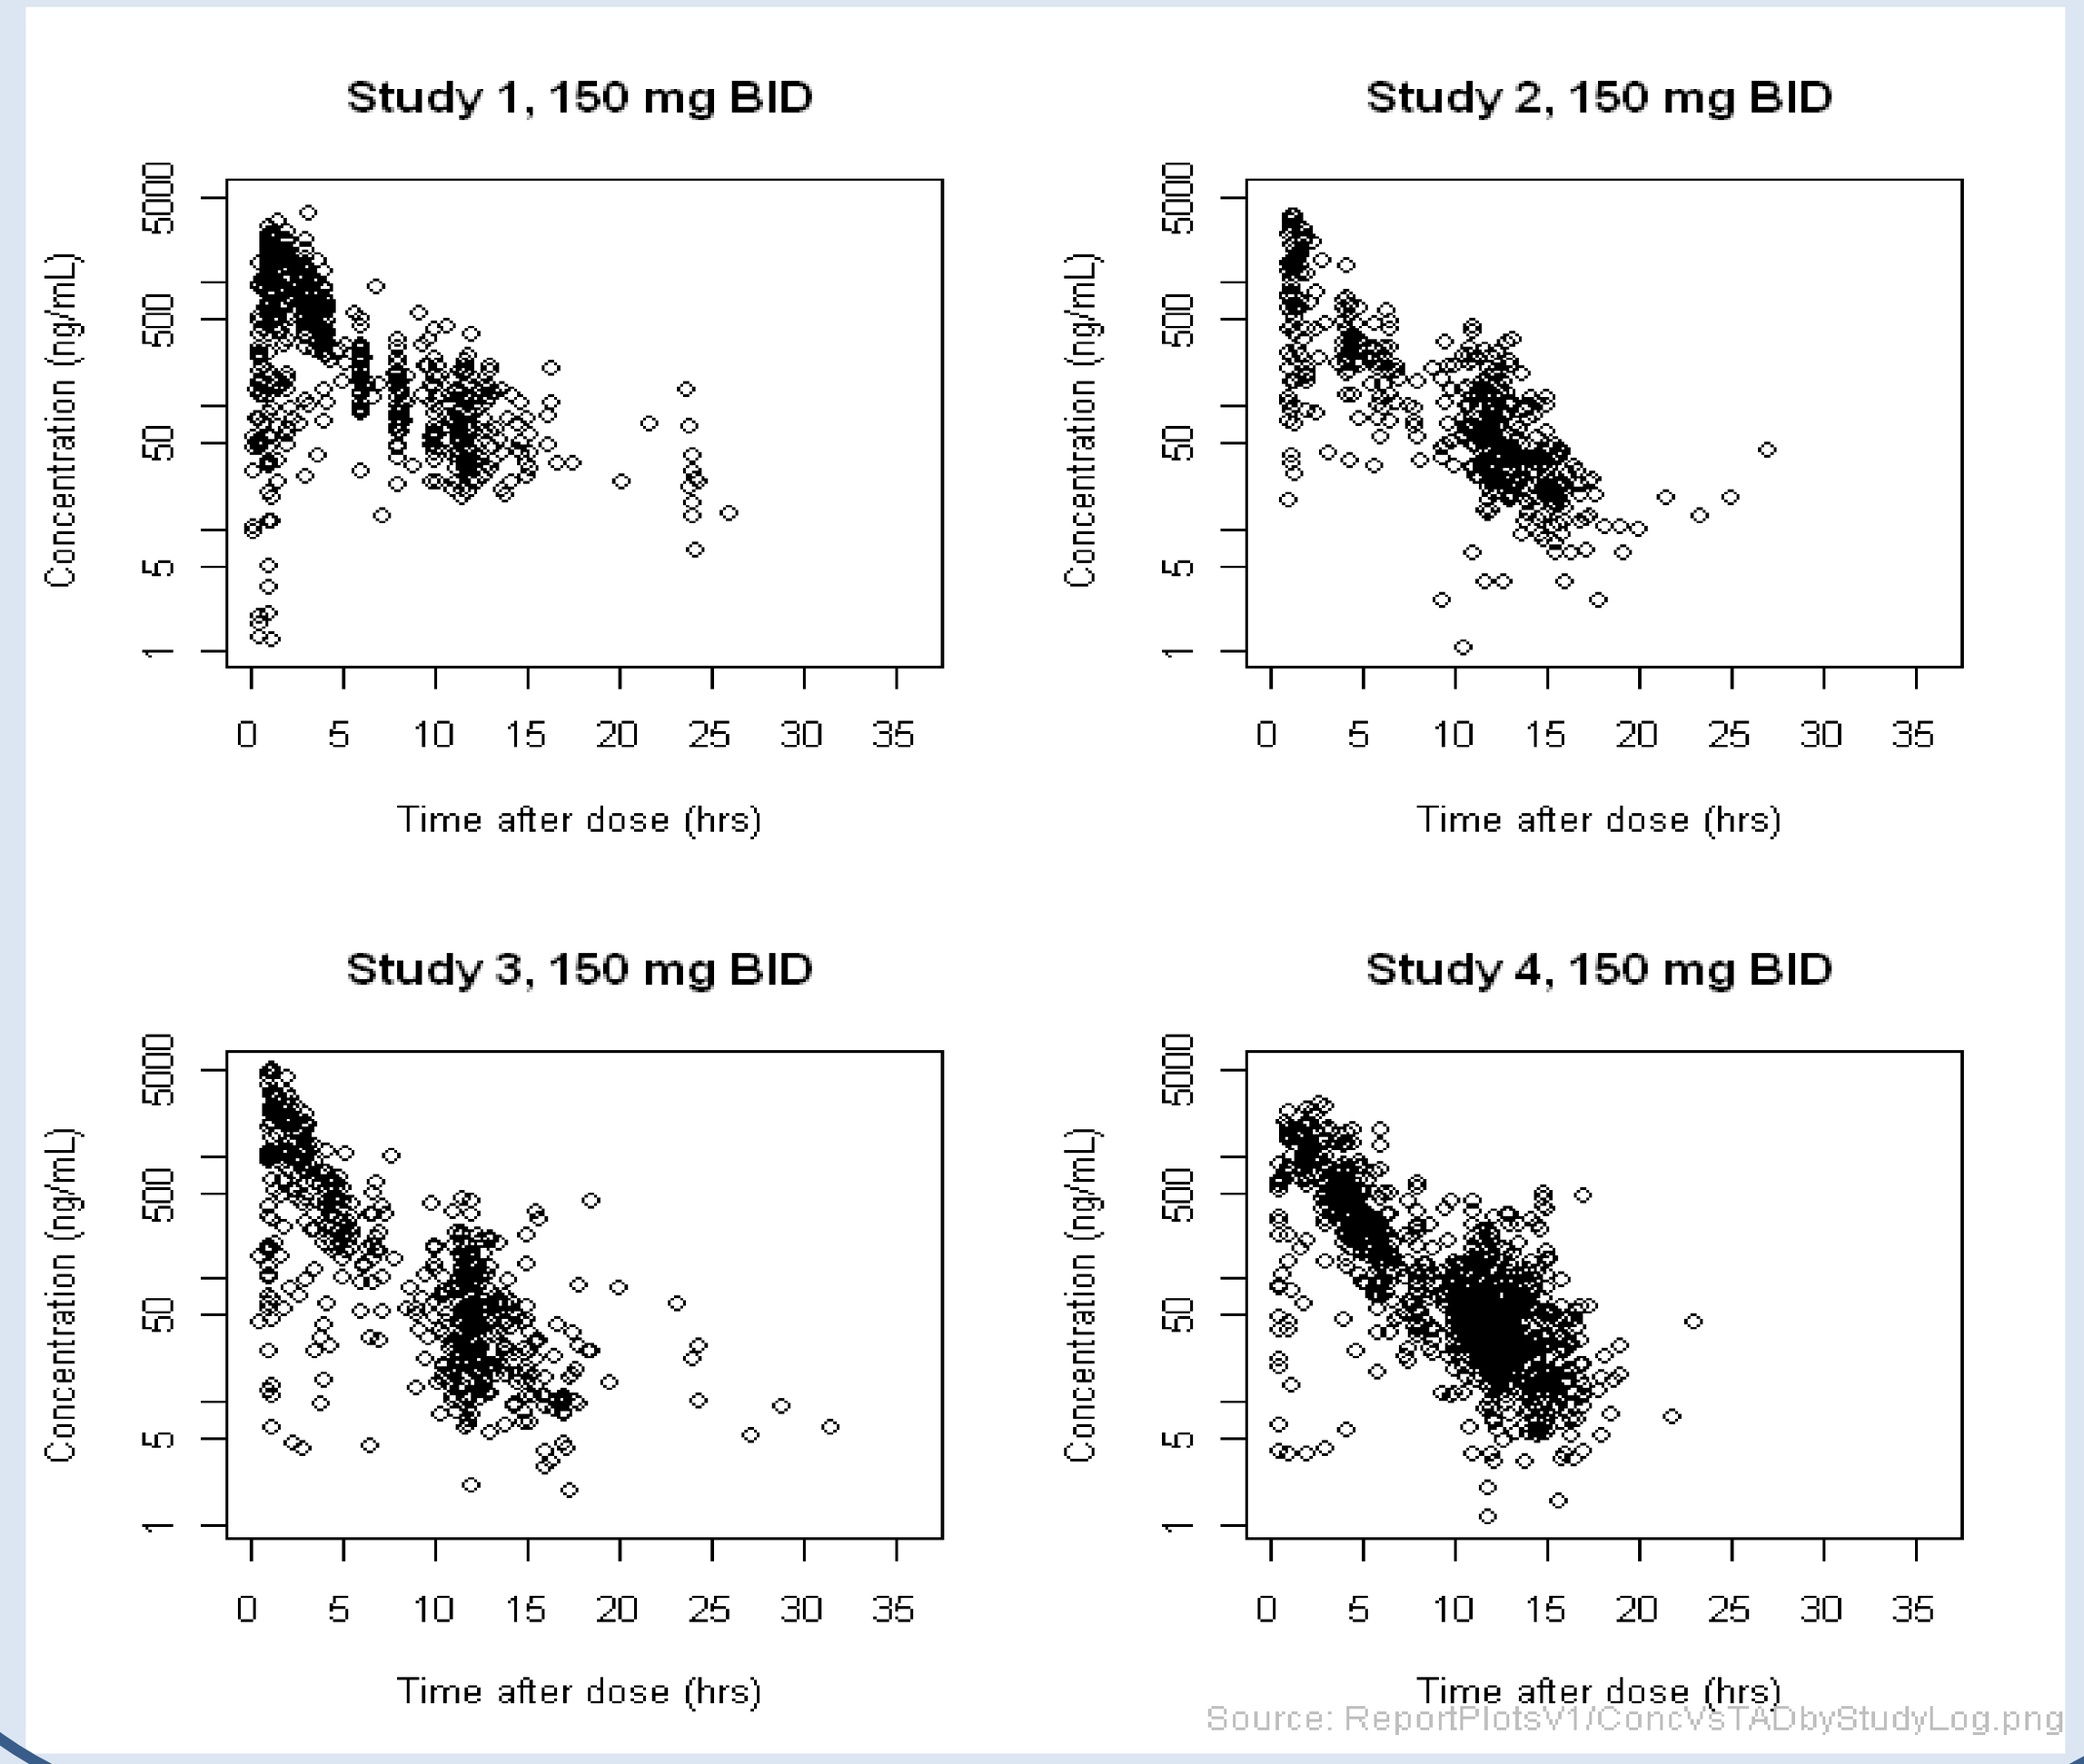
Study 1, 150 mg BID
Study 2, 150 mg BID
Study 3, 150 mg BID
Study 4, 150 mg BID
Concentration (ng/mL)
Time after dose (hrs)
Source: ReportPlotsV1/ConcVsTADbyStudyLog.png

Results – Cont.

- ❑ As shown in Figure 2, the effects of sex, body weight, and capsule shell were significant on CL/F and retained in the final model.
- ❑ Older age (categorized as ≤65, >65 to ≤ 75 years, and >75 years), mild and moderate renal impairment, mild hepatic impairment, CYP3A4 inhibitors, and CYP3A4 inducers were not significant predictors of oral clearance (CL/F) and the 95% CI were contained within 20% of null value.

Figure 2.

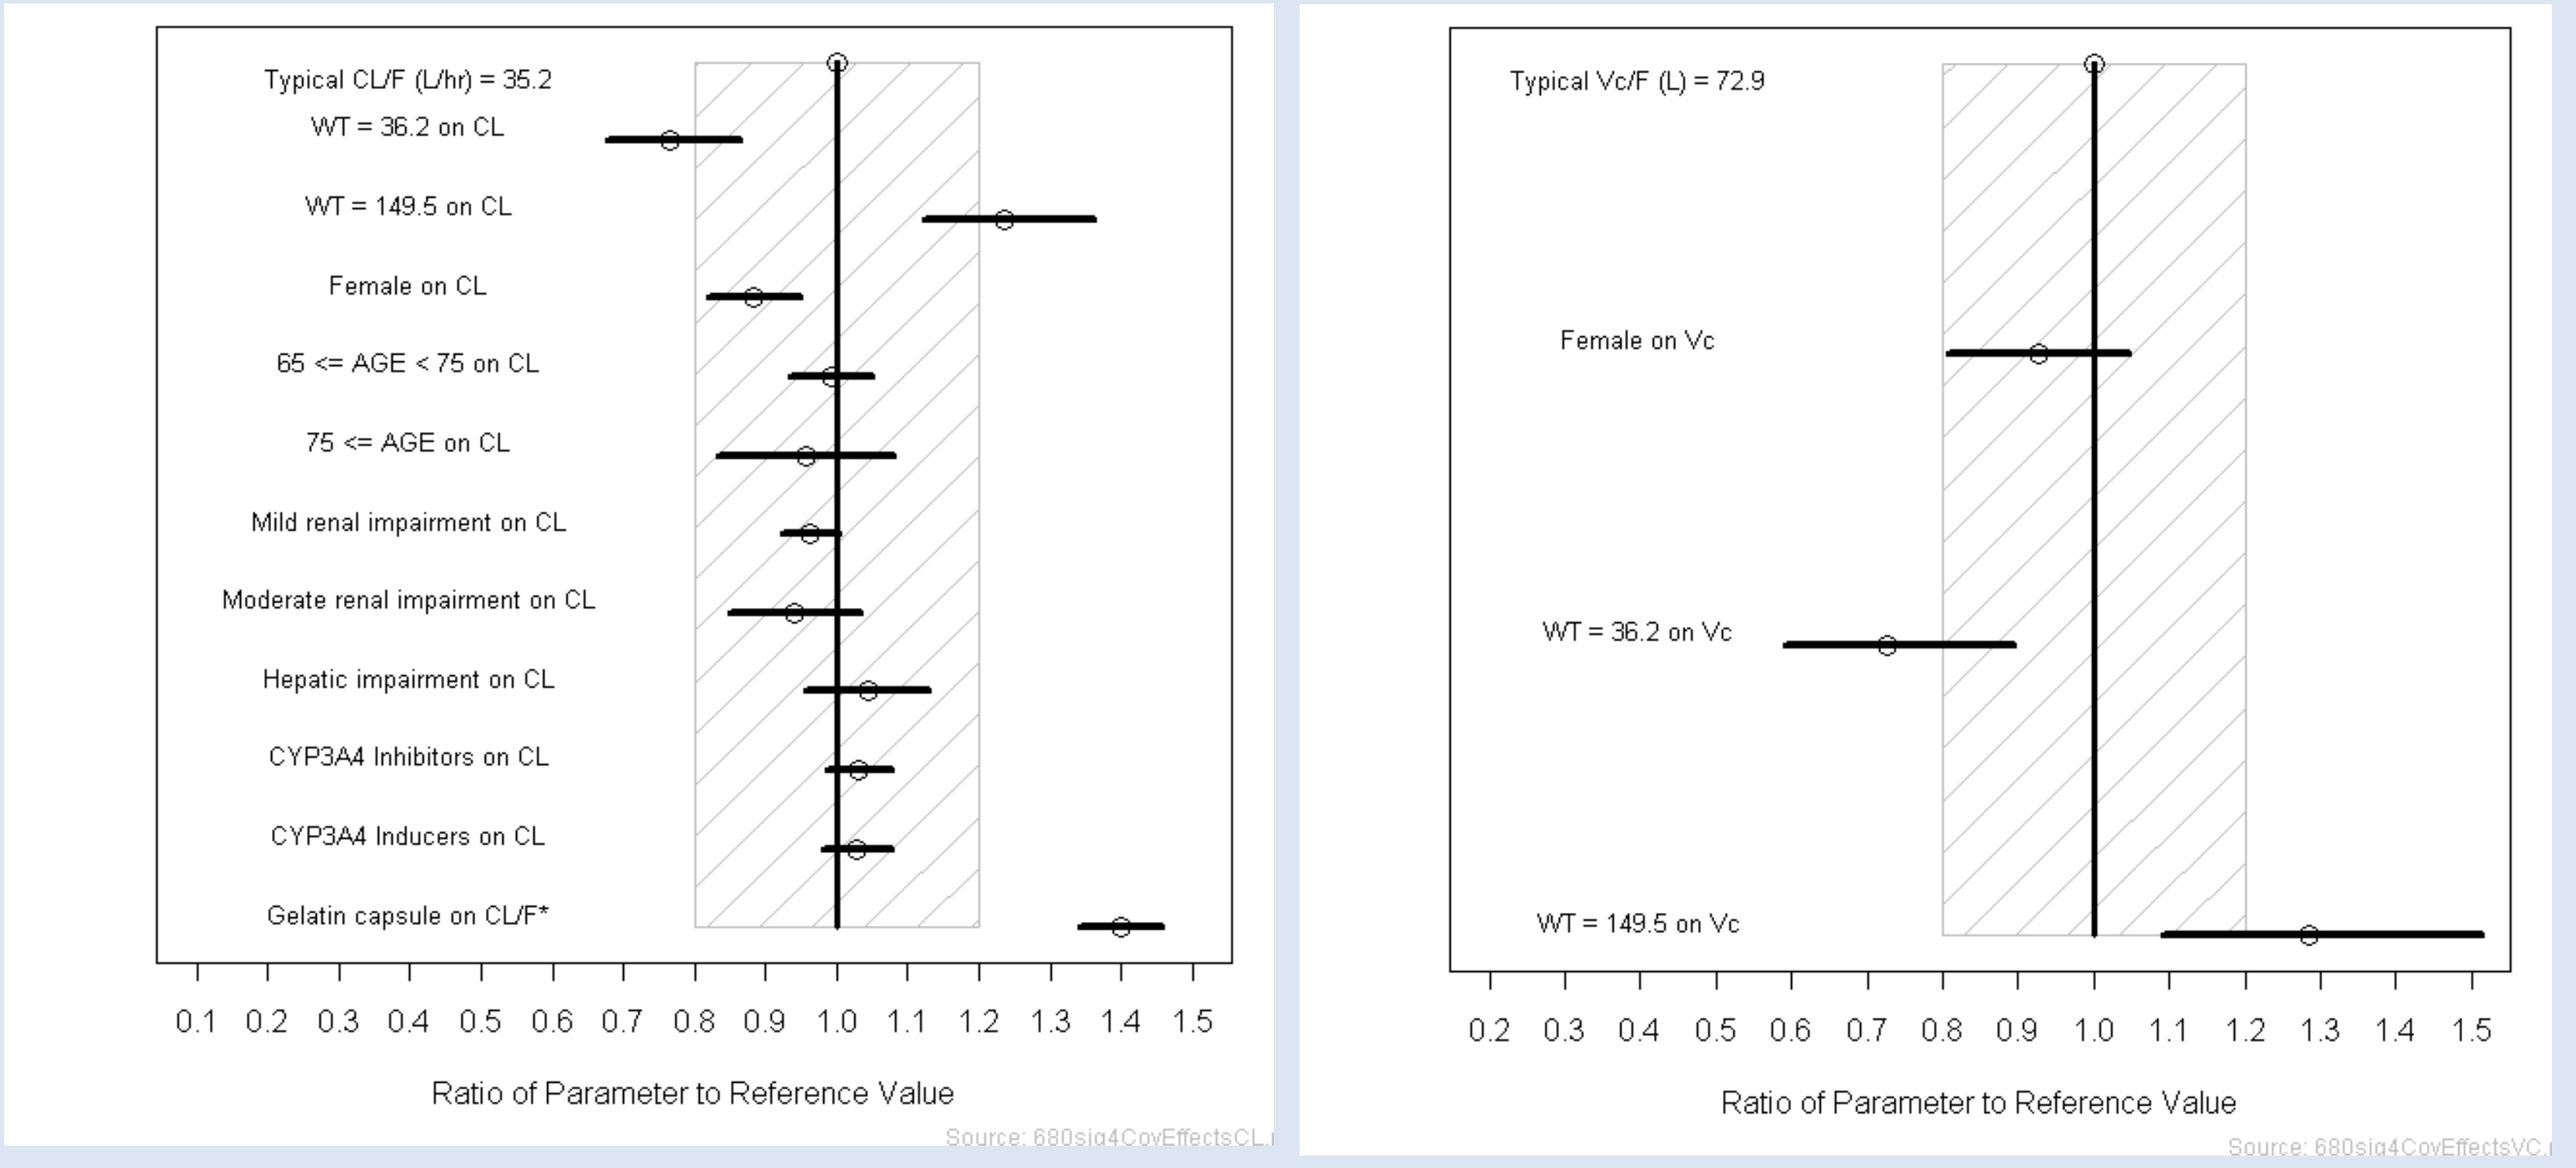
Typical CL/F (L/hr) = 35.2
WT = 36.2 on CL
WT = 149.5 on CL
Female on CL
65 <= AGE < 75 on CL
75 <= AGE on CL
Mild renal impairment on CL
Moderate renal impairment on CL
Hepatic impairment on CL
CYP3A4 Inhibitors on CL
CYP3A4 Inducers on CL
Gelatin capsule on CL/F*
Ratio of Parameter to Reference Value
Typical Vc/F (L) = 72.9
Female on Vc
WT = 36.2 on Vc
WT = 149.5 on Vc
Ratio of Parameter to Reference Value

- ❑ Final PK parameters are provided in Table 2.
- ❑ Based on T_{50} estimate of 67 hrs, steady-state is predicted to be achieved within 14 days of dosing (see Fig 3);
- ❑ Exposure decrease with repeat dosing with ratios of steady-state AUC(0-τ) and Cmax to single dose values were 0.50 and 0.88, respectively (see Fig 3);
- ❑ Predicted dose-proportionality shown in Fig 3.

Table 2

Parameter (descriptor)		Estimate (95% CI)	IIV (%CV)
CL_0/F (L/hr)	Non-Inducible CL/F	17.0 (15, 19)	58.6%
V_c/F (L)	Central volume	70.3 (62.7, 77.8)	53.0%
V_p/F (L)	Peripheral volume	154 (125, 183)	
Q/F (L/hr)	Distributional clearance	3.30 (2.82, 3.77)	99.0%
K_a (1/hr)	Absorption rate constant	1.88 (1.5, 2.25)	160%
Tlag (hr)	Lag time	0.482 (0.478, 0.486)	
$CL_{IND,SS}/F$ (L/hr)	Inducible CL/F	17.3 (16.2, 18.3)	
Alpha	Dose effect on $CL_{IND,ss}/F$	0.927 (0.842, 1.01)	
T_{50} (hr)	Half-life of induction	67.3 (47.2, 87.3)	
F_{GEL}	Relative F of gelatin vs. HPMC capsules	0.555 (0.488, 0.622)	
CL_{WT}	Weight effect on CL/F	0.331 (0.188, 0.474)	
CL_{SEX}	Sex effect on CL/F	0.914 (0.874, 0.954)	
Vc_{WT}	Weight effect on Vc/F	0.384 (0.15, 0.617)	
Q_{WT}	Weight effect on Q/F	1.22 (0.637, 1.8)	
Residual	Proportional error	0.343	53.0%
	Additive error (ng/mL)	0.292	SD=4.2

Figure 3.

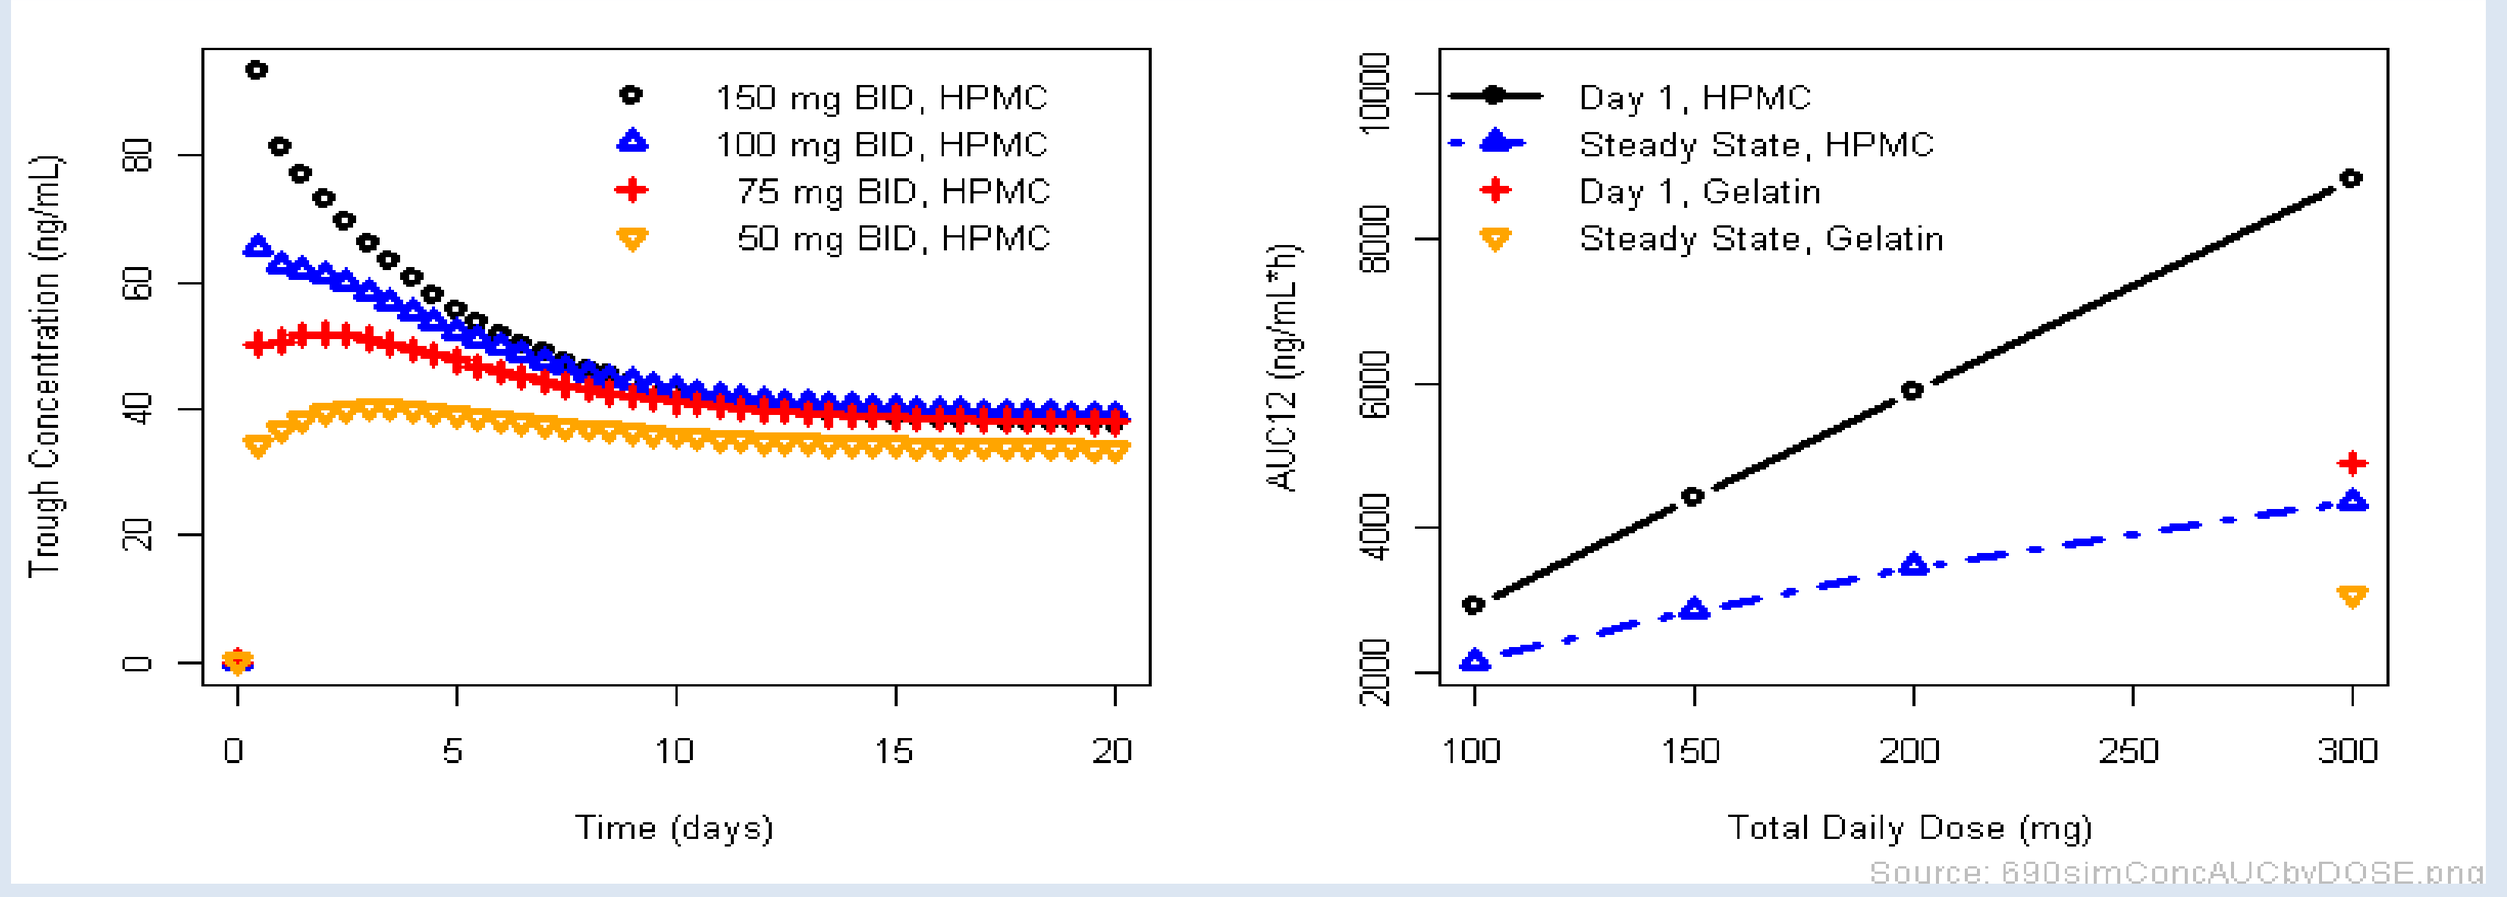
150 mg BID, HPMC
100 mg BID, HPMC
75 mg BID, HPMC
50 mg BID, HPMC
Trough Concentration (ng/mL)
Time (days)
Day 1, HPMC
Steady State, HPMC
Day 1, Gelatin
Steady State, Gelatin
AUC12 (ng/mL*h)
Total Daily Dose (mg)
Source: 690simConcAUCbyDOSE.png

Covariate Effects

- ❑ Capsule Type: Gelatin capsules have lower bioavailability compared to HPMC capsules. The ratio of HPMC over gelatin capsules after single dose is predicted to be 1.80 for both Cmax and AUC(0-τ). Following induction with repeat dosing, the ratio of HPMC to gelatin decreases to 1.66 and 1.42 for Cmax and AUC(0-τ), respectively. There was no difference in Ctrough between HPMC and gelatin capsules;
- ❑ Sex: CL/F was 8.6% lower in female subjects relative to male subjects; this difference was not considered clinically meaningful;
- ❑ Body Weight: Body weight influenced CL/F, Vc/F, and Q/F. PK parameters were predicted in a typical subject with low (50 kg) or high (140 kg) body weight and were shown to be within 20% of the value of a typical 80 kg subject. This difference was not considered clinically relevant.

Conclusions

- ❑ Dabrafenib PK adequately described by a two-compartment model with inducible oral clearance which increased with dose and with time with a half-life of induction of 67 hrs.
- ❑ IIV on PK parameters was high (53-160%).
- ❑ Following administration of 150 mg BID, the ratio of AUC(0-τ) of HPMC to gelatin capsules is predicted to be 1.80 after single dose and 1.42 at steady state.
- ❑ CL/F was 8.6% lower in female subjects relative to male subjects while CL/F, Vc/F and Q/F were influenced by body weight. These changes were small (within 20% of typical value) and not considered clinically relevant.
- ❑ Older age, mild and moderate renal impairment, mild hepatic impairment, and concomitant use of mild CYP3A4 inducers and inhibitors did not influence the PK or dabrafenib

All studies were funded by GlaxoSmithKline